Modules of genotypic variance reflect heterogeneity across TDP-43 proteinopathies

Barbara E Spencer, David J Irwin, Vivianna M Van Deerlin, Eddie B Lee, Lauren Elman,
Colin Quinn, Murray Grossman, David A Wolk, and Corey T McMillan
University of Pennsylvania, Philadelphia, PA, USA

Background

TAR DNA-binding protein (TDP-43) proteinopathies yield a variety of neurodegenerative conditions including amyotrophic lateral sclerosis (ALS), frontotemporal lobar degeneration (FTLD), and hippocampal sclerosis of aging (HS-Aging).

The contribution of both shared and disparate single nucleotide polymorphisms (SNPs) to phenotypic presentations is unclear.

We hypothesize that shared genetic variation may drive susceptibility across diseases while disparate genetic variation may contribute to our understanding of disease-specific drivers.

Methods

• Using existing GWAS summary statistics with novel bioinformatics strategies, we aimed to identify modules of SNPs that drive the cognitive/behavioral and/or neuromuscular presentations of TDP-43 proteinopathies, identify the biological pathways associated with these module SNPs, and test for associations between module-specific polygenic risk and phenotypic heterogeneity.

• We compared the relative associations of GWAS summary statistics for ALS,[1] FTLD-TDP,[2] and HS-Aging[3] using weighted correlation analysis[4] to identify data-driven modules of highly correlated SNPs.

GWAS assume that SNPs associated with a phenotype are relevant to that disease.

Here, weighted correlation analysis uses the patterns of associations of SNPs with ALS, FTLD-TDP, and HS-Aging to identify correlations between SNPs and construct modules based on those correlations.

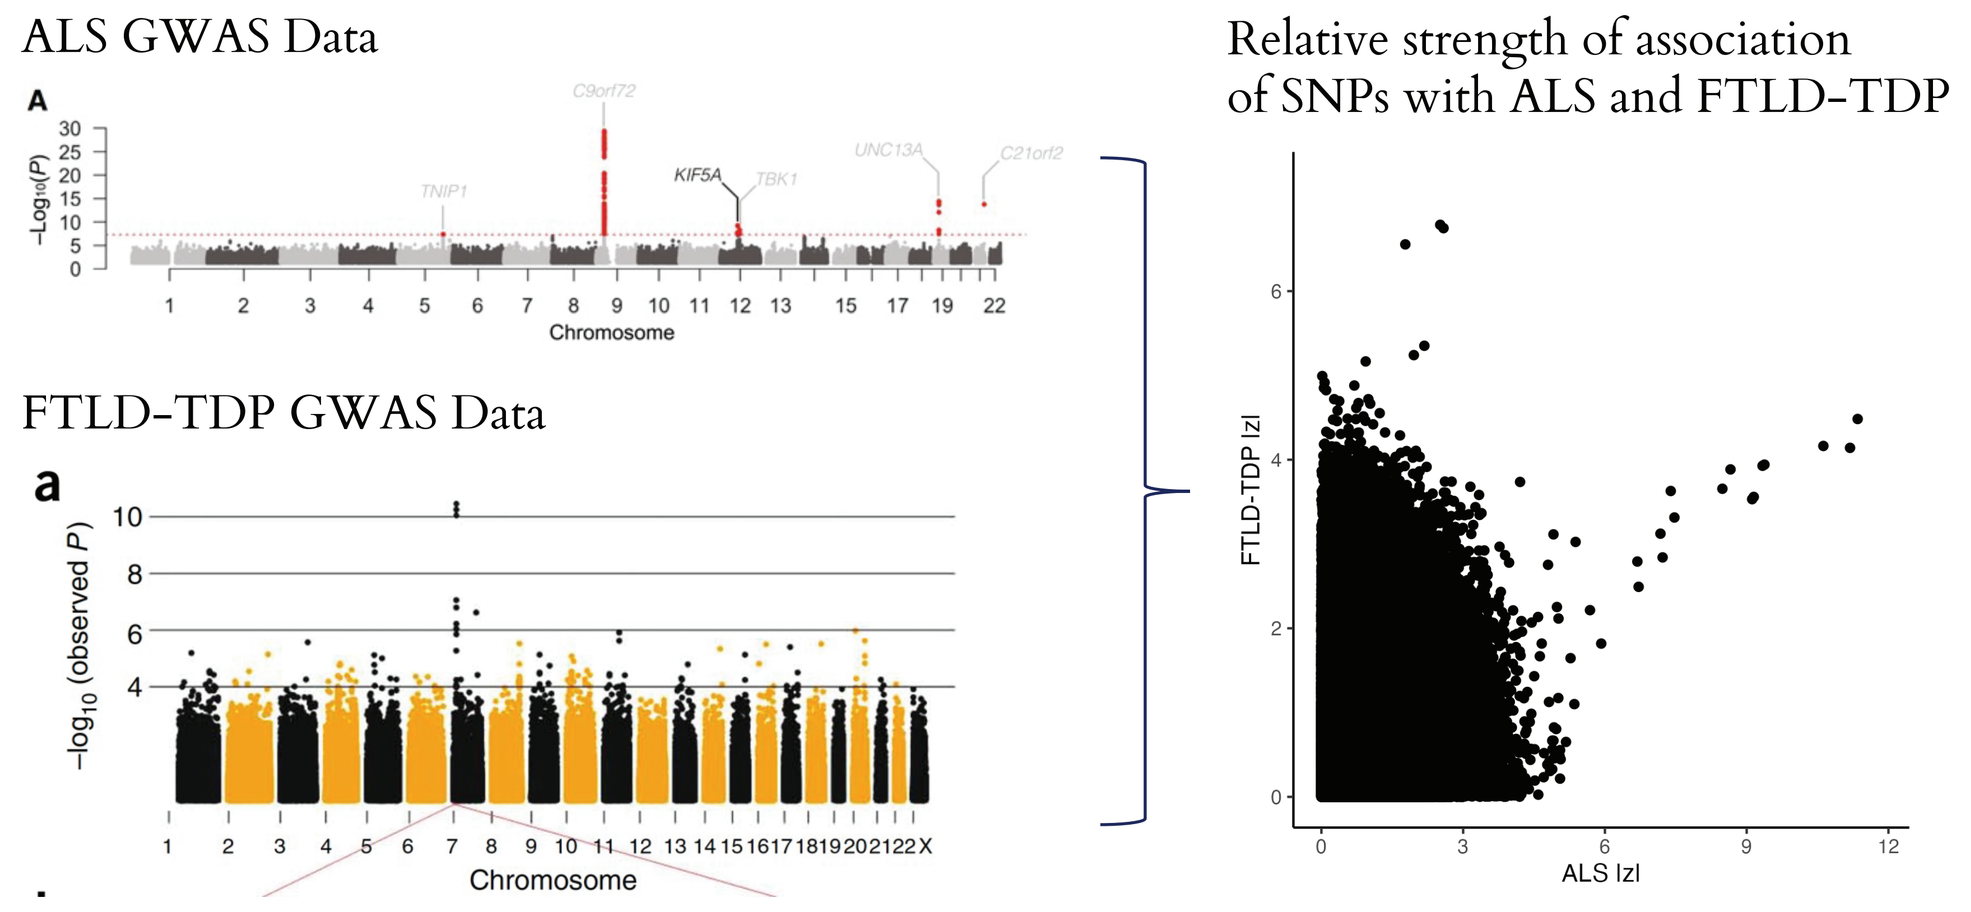
ALS GWAS Data
FTLD-TDP GWAS Data
Chromosome
Relative strength of association of SNPs with ALS and FTLD-TDP
FTLD-TDP |z|
ALS |z|

• Module SNPs were mapped to genes with biomaRt.[5] A gene ontology enrichment analysis for biological processes was performed separately for each identified module with g:Profiler.[6]

• Polygenic risk scores (PRS) were constructed using module SNPs and tested for association with diagnosis in a sample of cases that met either neuropathological or genetic criteria.

Results

Examining genotypes revealed modules of both shared and disparate correlated SNPs across phenotypes associated with TDP-43 proteinopathies which have biologically plausible pathway associations.

Weighted correlation analysis identified 5 modules of correlated SNPs associated with either a single GWAS phenotype or phenotypic pairs.

This approach used the patterns of associations of SNPs with each GWAS phenotype to identify correlations between SNPs and construct modules.

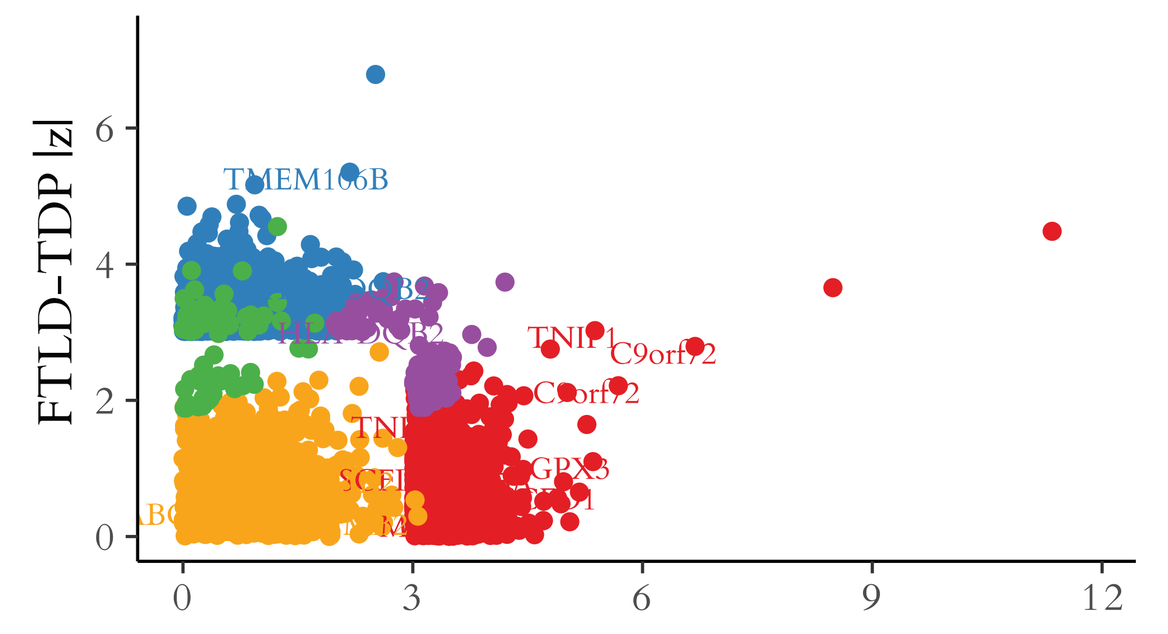
FTLD-TDP |z|
TMEM106B
C9orf72

• 494,417 SNPs were commonly genotyped across ALS, FTLD-TDP, and HS-Aging GWAS summary statistics.

• The top 1% of these SNPs with the strongest associations to any GWAS phenotype (by |z|, n= 4,945) were selected for analysis.

• After modules were identified, SNPs were pruned within modules using an R^2 threshold of 0.3 with LDlinkR.

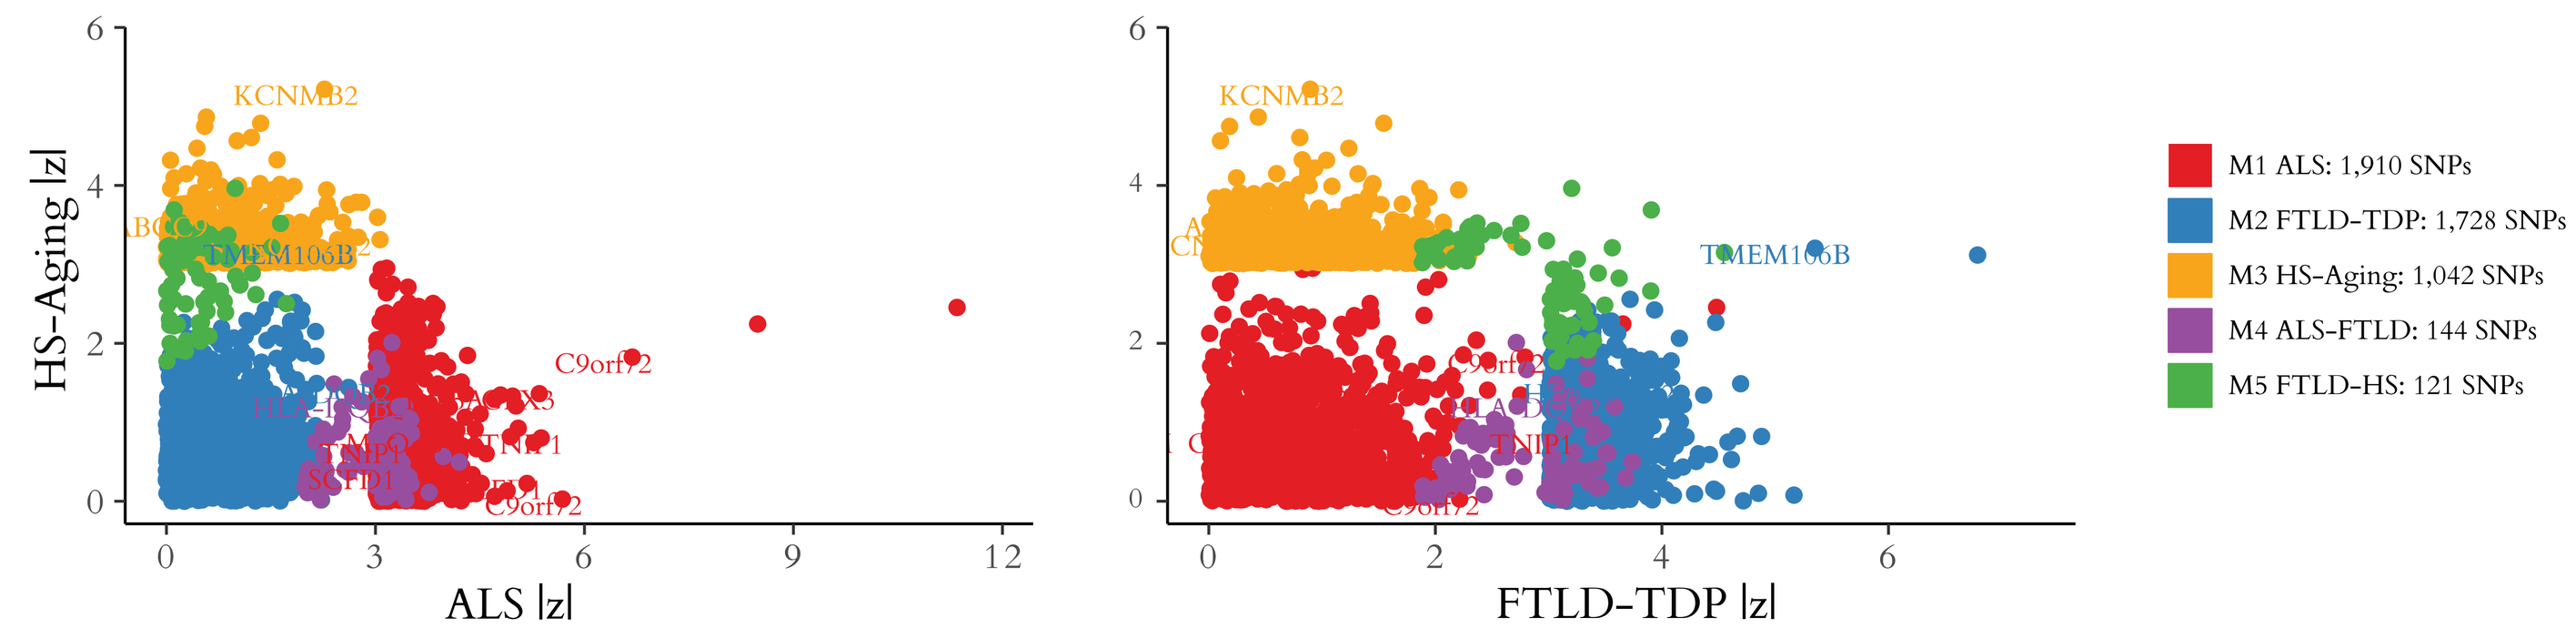
HS-Aging |z|
ALS |z|
FTLD-TDP |z|
KCNMB2
TMEM106B
C9orf72
M1 ALS: 1,910 SNPs
M2 FTLD-TDP: 1,728 SNPs
M3 HS-Aging: 1,042 SNPs
M4 ALS-FTLD: 144 SNPs
M5 FTLD-HS: 121 SNPs

• Modules were associated with either a single GWAS phenotype: ALS (M1), FTLD-TDP (M2), and HS-Aging (M3), or phenotypic pairs: ALS-FTLD (M4) and FTLD-HS (M5).

Gene ontology enrichment analysis for biological processes revealed pathways relevant to nervous system development.

Notably, M1 ALS uniquely includes pathways related to spinal cord, while pathways related to neurogenesis were uniquely observed in M3 HS-Aging.

• Module SNPs were mapped to genes with biomaRt.

• A gene ontology enrichment analysis for biological processes was performed for each module with g:Profiler.

• Fold enrichment was calculated using an expected value based on matching the proportion of genes in a particular term in the human genome reference list to the number of genes provided.

• The top 20 overrepresented pathways by fold enrichment that met FDR p<.05 are displayed for each module, with relevant themes highlighted for each module.

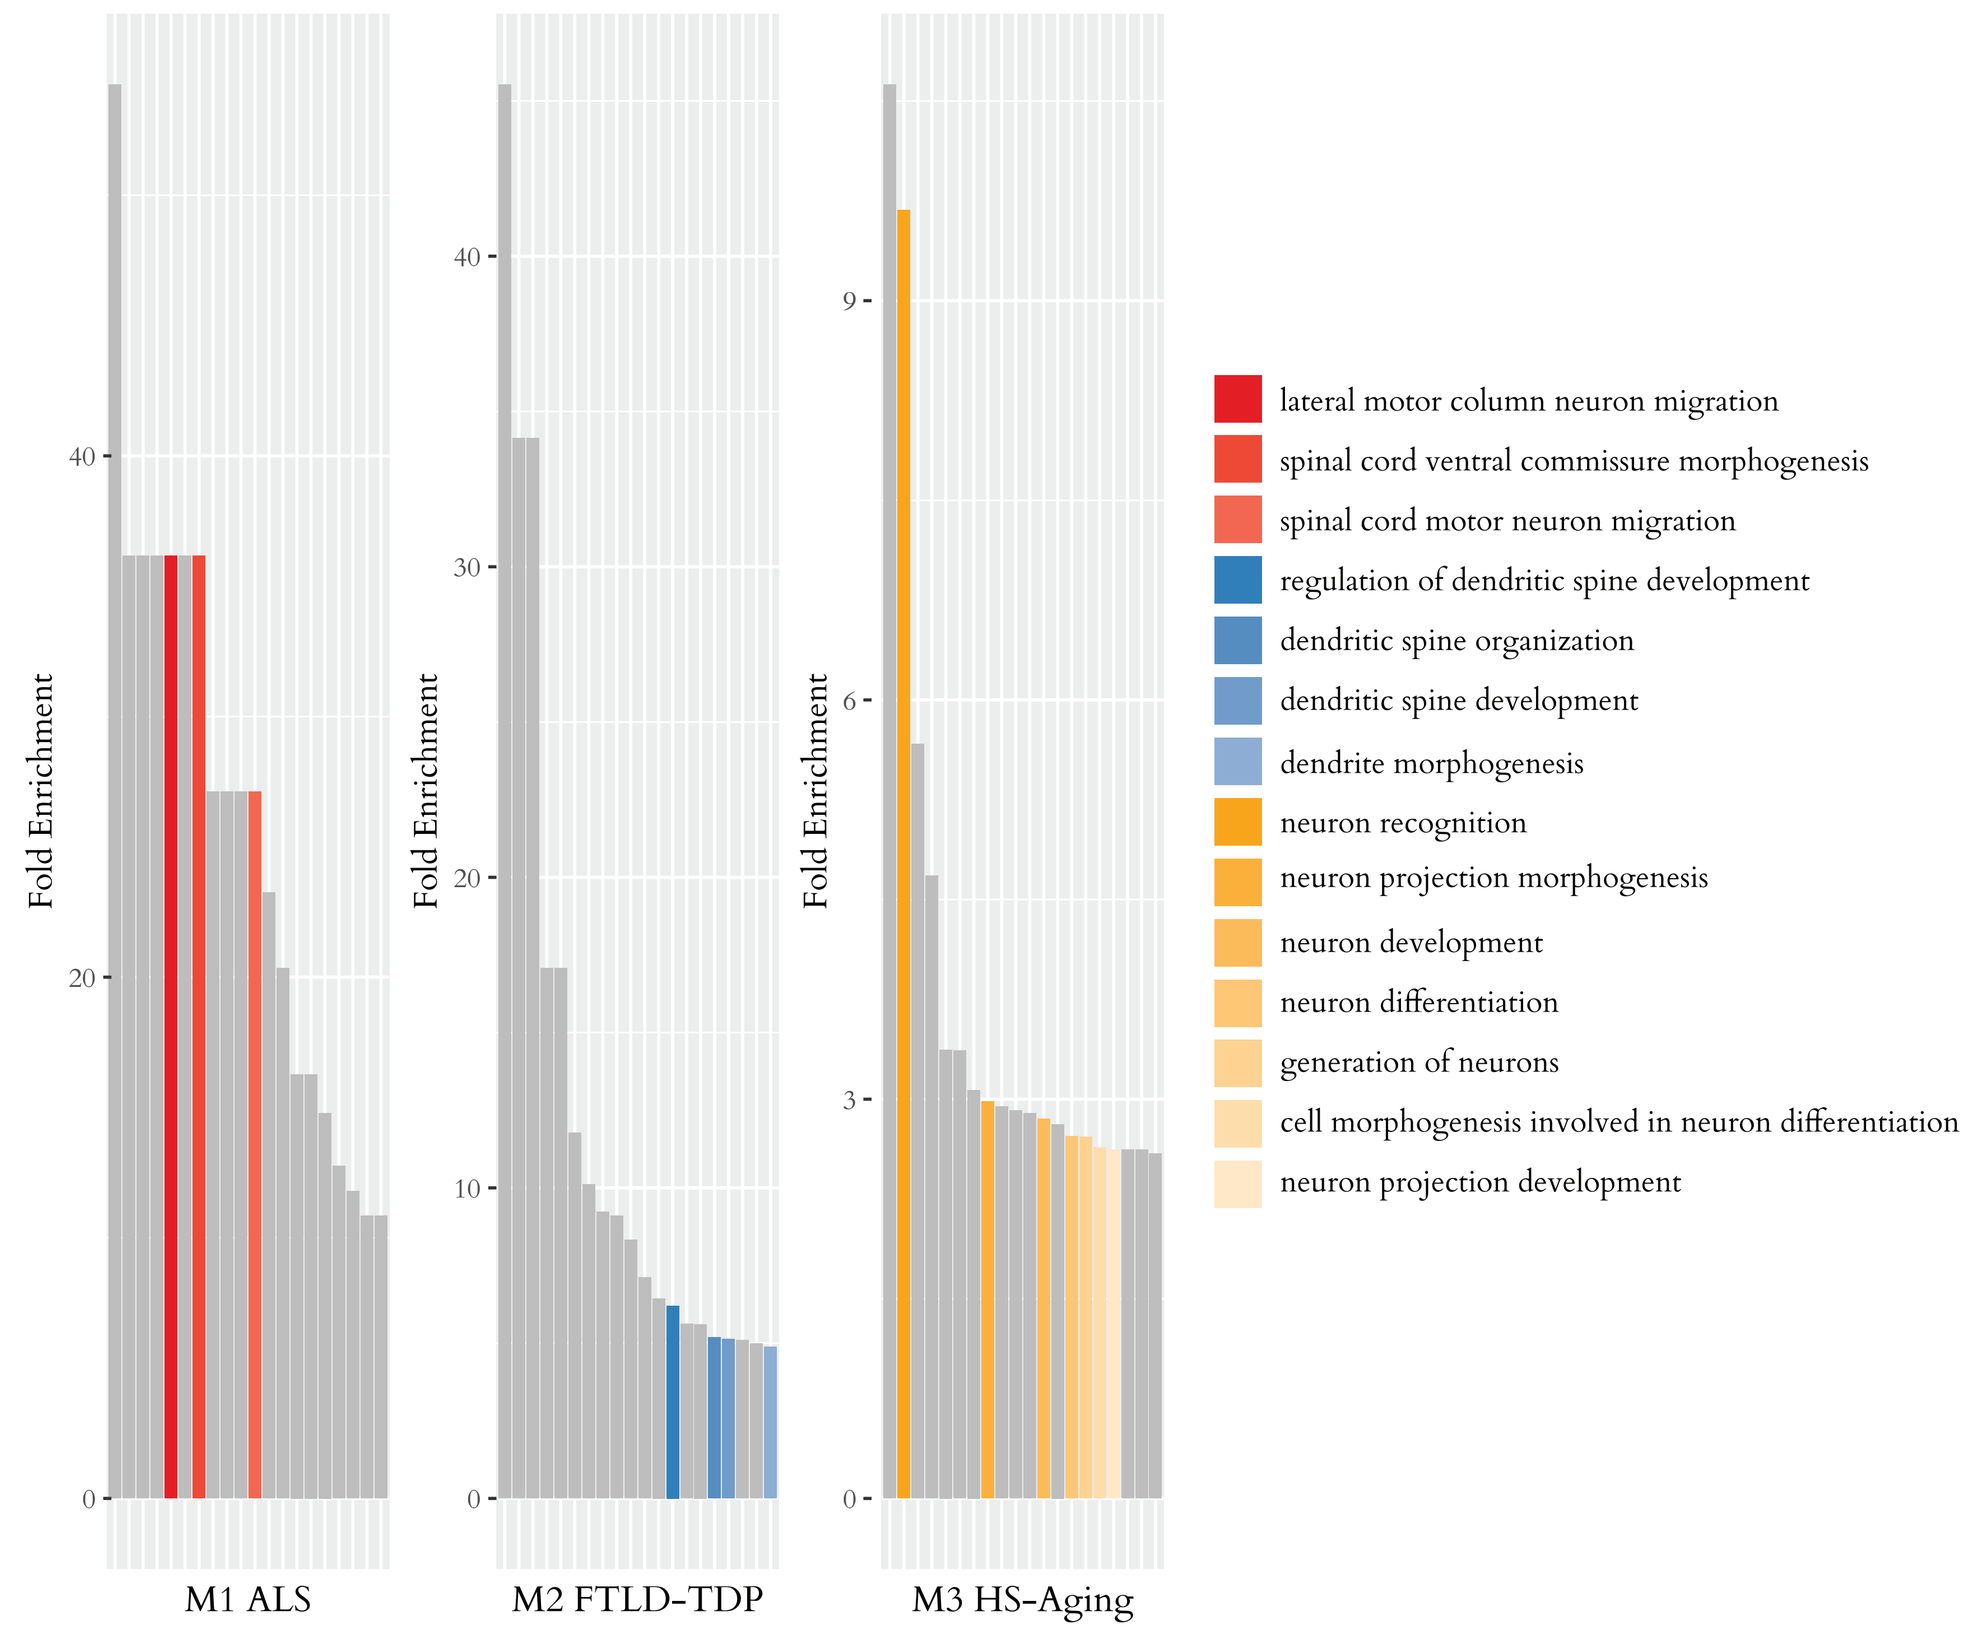
Fold Enrichment
M1 ALS
M2 FTLD-TDP
M3 HS-Aging
lateral motor column neuron migration
spinal cord ventral commissure morphogenesis
spinal cord motor neuron migration
regulation of dendritic spine development
dendritic spine organization
dendritic spine development
dendrite morphogenesis
neuron recognition
neuron projection morphogenesis
neuron development
neuron differentiation
generation of neurons
cell morphogenesis involved in neuron differentiation
neuron projection development

Higher M2 FTLD-TDP than M1 ALS polygenic risk was observed in ALS-FTD and FTLD-TDP cases compared to ALS cases.

	ALS	ALS-FTD	FTLD-TDP
n	172	54	170
Sex, Male (%)	103 (59.9)	26 (48.1)	92 (54.1)
Education (mean (SD))	15.06 (3.27)	14.26 (2.97)	15.47 (3.17)
Deceased (%)	168 (97.7)	54 (100.0)	143 (84.1)
Age at Death (mean (SD))	63.19 (10.40)	61.19 (8.64)	69.14 (9.53)
Race (%)			
Asian	0 (0.0)	1 (1.9)	1 (0.6)
Black or African American	5 (2.9)	1 (1.9)	2 (1.2)
More than One Race	0 (0.0)	0 (0.0)	4 (2.4)
White	163 (94.8)	50 (92.6)	159 (93.5)
Unknown or Not Reported	4 (2.3)	2 (3.7)	4 (2.4)
Ethnicity (%)			
Hispanic or Latino	3 (1.7)	0 (0.0)	2 (1.2)
Not Hispanic or Latino	163 (94.8)	53 (98.1)	164 (96.5)
Unknown or Not Reported	6 (3.5)	1 (1.9)	4 (2.4)
Mutation (%)			
No Known Mutation	112 (65.1)	15 (27.8)	65 (38.2)
Mutation Carrier	60 (34.9)	39 (72.2)	105 (61.8)
Prior GWAS Case (%)	62 (36.0)	20 (37.0)	50 (29.4)

• To address whether module-specific polygenic risk was associated with phenotypic heterogeneity, we constructed module-specific PRS for M1 ALS and M2 FTLD-TDP.

• The PRS is the non-weighted sum of risk alleles per SNP per module for each individual, centered using TOPMed global population allele frequencies and mean scaled.

• The difference between the M2 FTLD-TDP and M1 ALS PRS was evaluated to assess relative risk.

• Multinomial logistic regression models were used to examine the relationship between PRS and diagnostic group, controlling for age at death and sex.

• Higher M2 FTLD-TDP than M1 ALS PRS was observed in both ALS-FTD and FTLD-TDP cases compared to ALS cases (p<.01). As some cases were included in prior GWAS for ALS and FTLD-TDP, we confirmed that this difference held in the subset of independent ALS and FTLD-TDP cases with a t-test (p=.03).

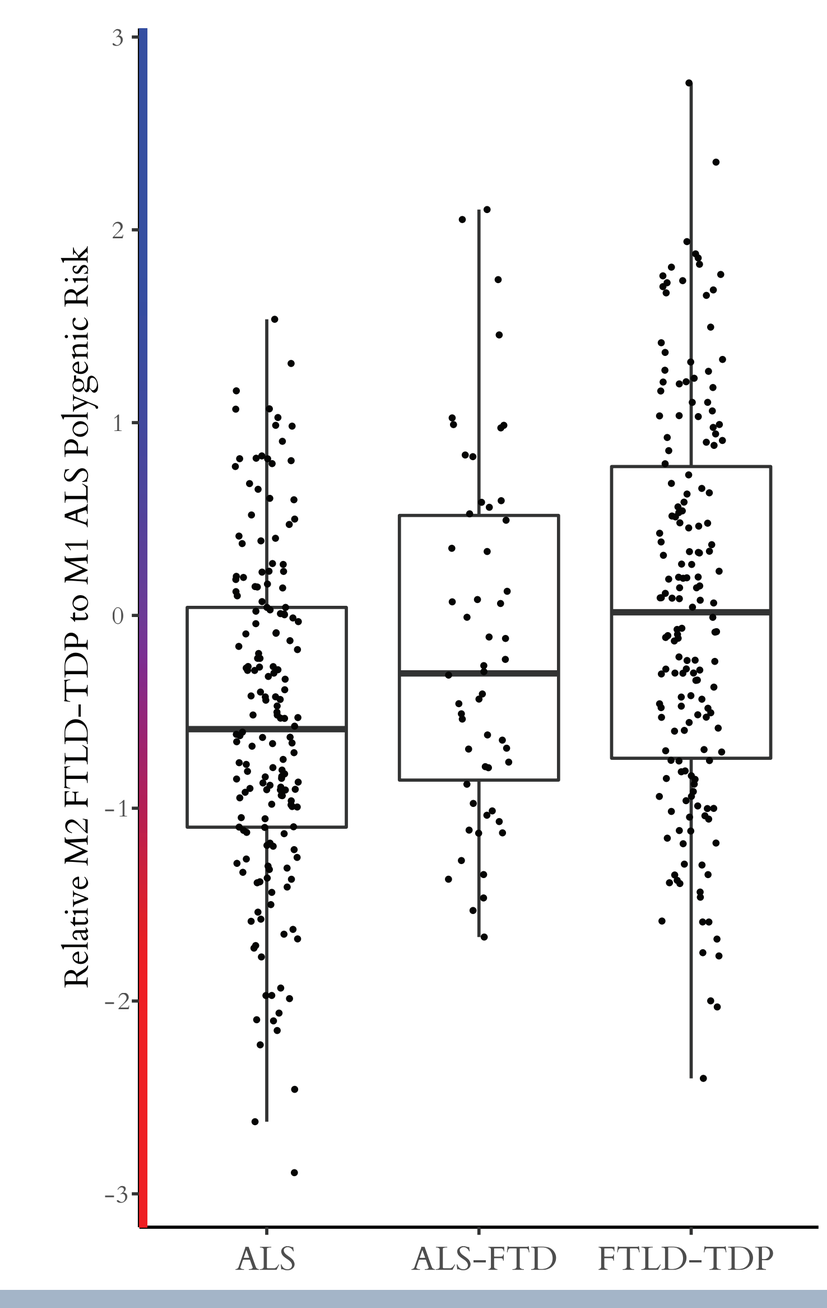
Relative M2 FTLD-TDP to M1 ALS Polygenic Risk
ALS
ALS-FTD
FTLD-TDP

Conclusion

We suggest that genetic variation across TDP-43 proteinopathies may contribute to individual-level presentations of these syndromes.

Identifying disparate risk alleles may reveal disease-specific anatomic and/or cellular vulnerabilities that drive ALS and FTLD-TDP.

References

1. Nicolas et al. Genome-wide Analyses Identify KIF5A as a Novel ALS Gene. Neuron. 2018 Mar;97(6):1268-1283.e6.
2. Van Deerlin et al. Common variants at 7p21 are associated with frontotemporal lobar degeneration with TDP-43 inclusions. Nat Genet. 2010 Mar;42(3):234–239.
3. Beecham et al. Genome-Wide Association Meta-analysis of Neuropathologic Features of Alzheimer's Disease and Related Dementias. PLoS Genet. 2014 Sep 4;10(9):e1004606.
4. Langfelder, Horvath. WGCNA: an R package for weighted correlation network analysis. BMC Bioinformatics. 2008 Dec 29;9(1):559.
5. Smedley et al. The BioMart community portal: an innovative alternative to large, centralized data repositories. Nucleic Acids Research. 2015 Apr 20;43(W1):W589–W598.
6. Raudvere et al. g: Profiler: a web server for functional enrichment analysis and conversions of gene lists. Nucleic Acids Research. 2019 May 8;47(W1):W191-W198.